Etrasimod (APD334), an Oral, Next-Generation Sphingosine-1-Phosphate Receptor Modulator Inhibits the Development of Colitis in Lymphoid-Null Mice Injected with Colitogenic CD4+ T Cells

Author: John W. Adams[1], Michelle Solomon[1], Dominic Behan, Chris Carroll[2], Karin Lehmann-Bruinsma[2], Hussien Al-Shamma[2]
Affiliations: [1]Arena Pharmaceuticals and [2]Beacon Discovery, San Diego, CA, USA

Abstract #6337

Introduction

Etrasimod (APD334) is a next-generation oral sphingosine-1-phosphate (S1P) receptor modulator with an optimized S1P receptor activity profile compared to first generation S1P drugs, currently in Phase 2 clinical development for the treatment of ulcerative colitis (UC). Preventing the circulation of effector lymphocytes from peripheral lymphoid organs by S1P1 modulators, including fingolimod (FTY-720), has been shown to be effective in preclinical models of inflammatory bowel diseases[1-5]. We studied the effects of etrasimod on experimental colitis in the CD4+CD45RB^{high} T cell adoptive transfer model. Our findings suggest that etrasimod is a potent immunomodulating drug that may be effective in treating T cell dependent inflammatory bowel diseases including UC.

Methods

Female SCID mice (9 weeks) were injected intraperitoneally with 5x10^{5} CD4+CD45RB^{high} cells isolated from the spleens of BALB/c female donor mice. Unsorted, enriched T cells were also injected into one group of mice to serve as control. All animals were dosed (etrasimod 1, 3 mg/kg p.o., FTY720 1mg/kg p.o.) once daily beginning the day of adoptive transfer until the day before tissue harvest. At study termination animals were euthanized and tissues were collected for histological and quatitiative PCR (qPCR) analyses. For qPCR, gene expression was normalized to glyceraldehyde phosphodehydrogenase (GAPDH) levels, then analyzed using the $\Delta\Delta$ Ct method and reported as fold/control. One-way ANOVA followed by Dunnett's multiple comparisons test was used to compare treatment vs. vehicle control.

Results

Approximately 3 weeks after transfer, SCID mice that received CD4+ CD45RB^{high} T cells progressively developed symptoms of colitis, including weight loss and loose mucinous stool, compared with mice that received unsorted CD4+ T cells. These symptoms were significantly reduced in response to treatment with etrasimod as observed in both in life and post mortem analyses summarized in Figures 1-4.

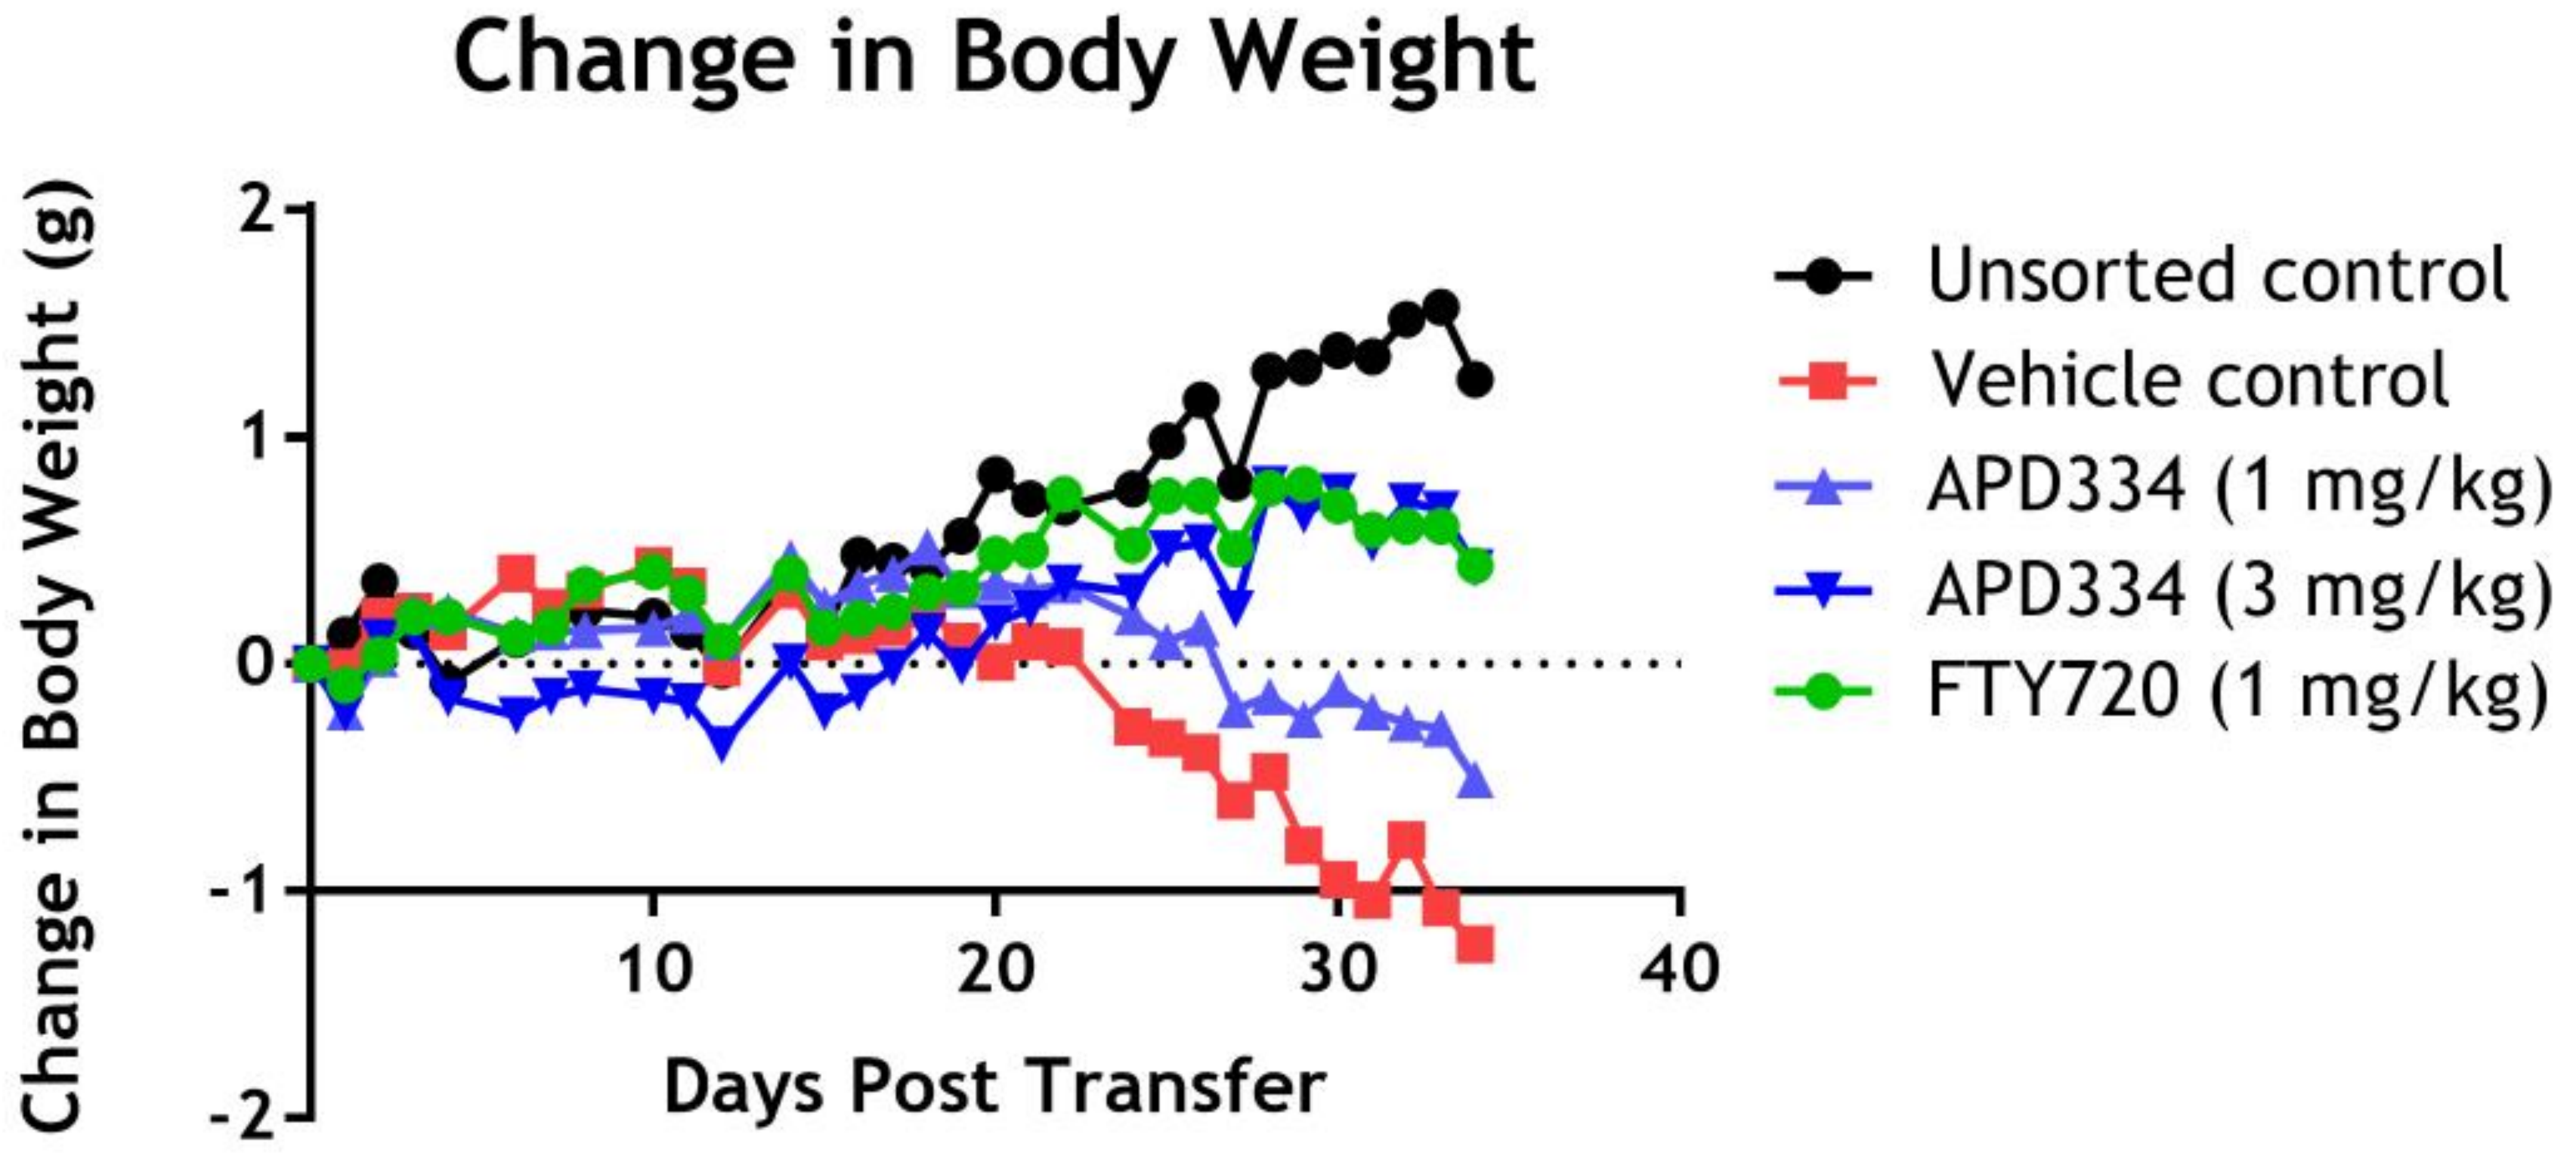
Change in Body Weight
Change in Body Weight (g)
Days Post Transfer
Unsorted control
Vehicle control
APD334 (1 mg/kg)
APD334 (3 mg/kg)
FTY720 (1 mg/kg)

Figure 1 *Treatment with etrasimod or FTY-720 inhibits weight loss associated with disease onset in SCID mice receiving CD4+ colitogenic T cells.*

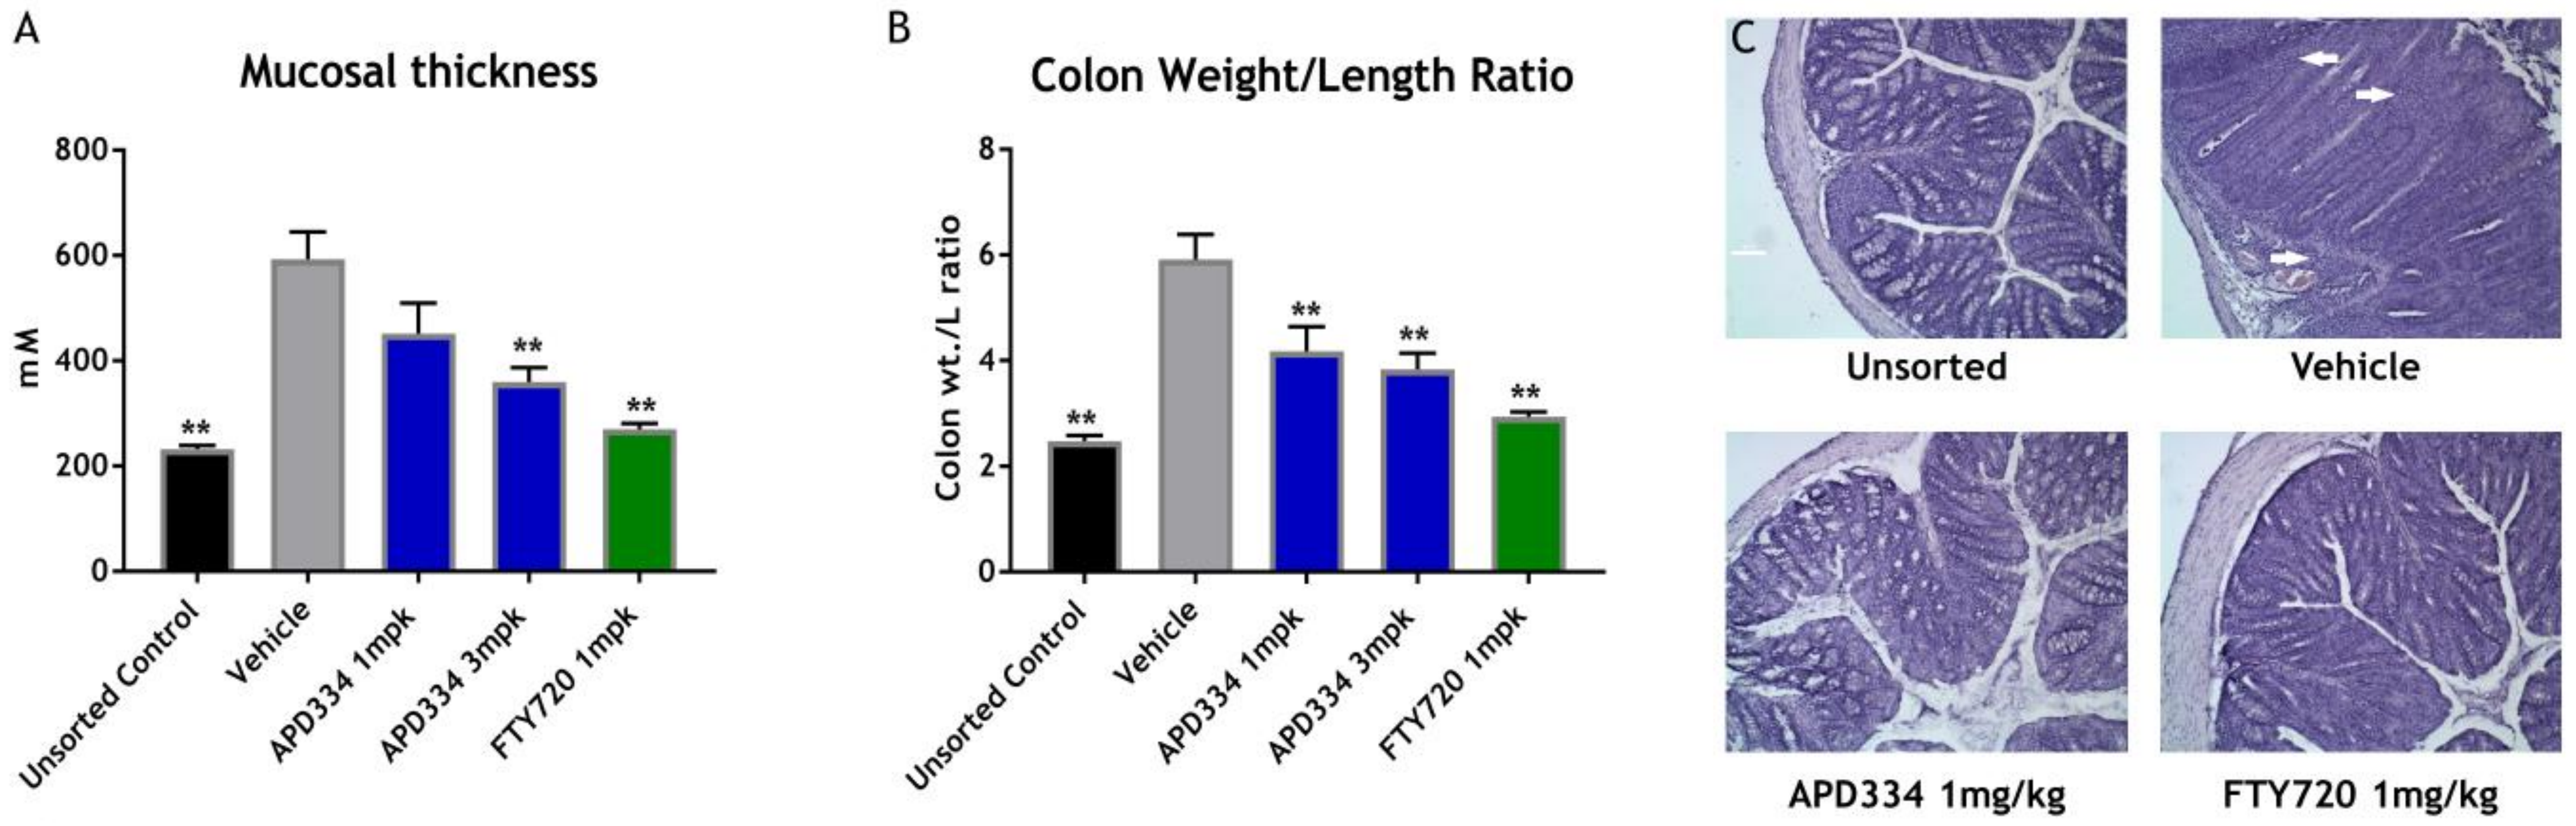
A
Mucosal thickness
mM
B
Colon Weight/Length Ratio
Colon wt./L ratio
Unsorted Control
Vehicle
APD334 1mpk
APD334 3mpk
FTY720 1mpk
C
Unsorted
Vehicle
APD334 1mg/kg
FTY720 1mg/kg

Figure 2 *Etrasimod inhibits the increase in histological markers of disease. A) Etrasimod inhibits increases in mucosal thickening. B) Etrasimod reduces colon weight to length ratio, and (C) Photomicrographs of H and E stained colon cross sections suggest that etrasimod decreases inflammatory cell infiltration (arrows) in the colons of mice transferred with colitogenic T cells.*

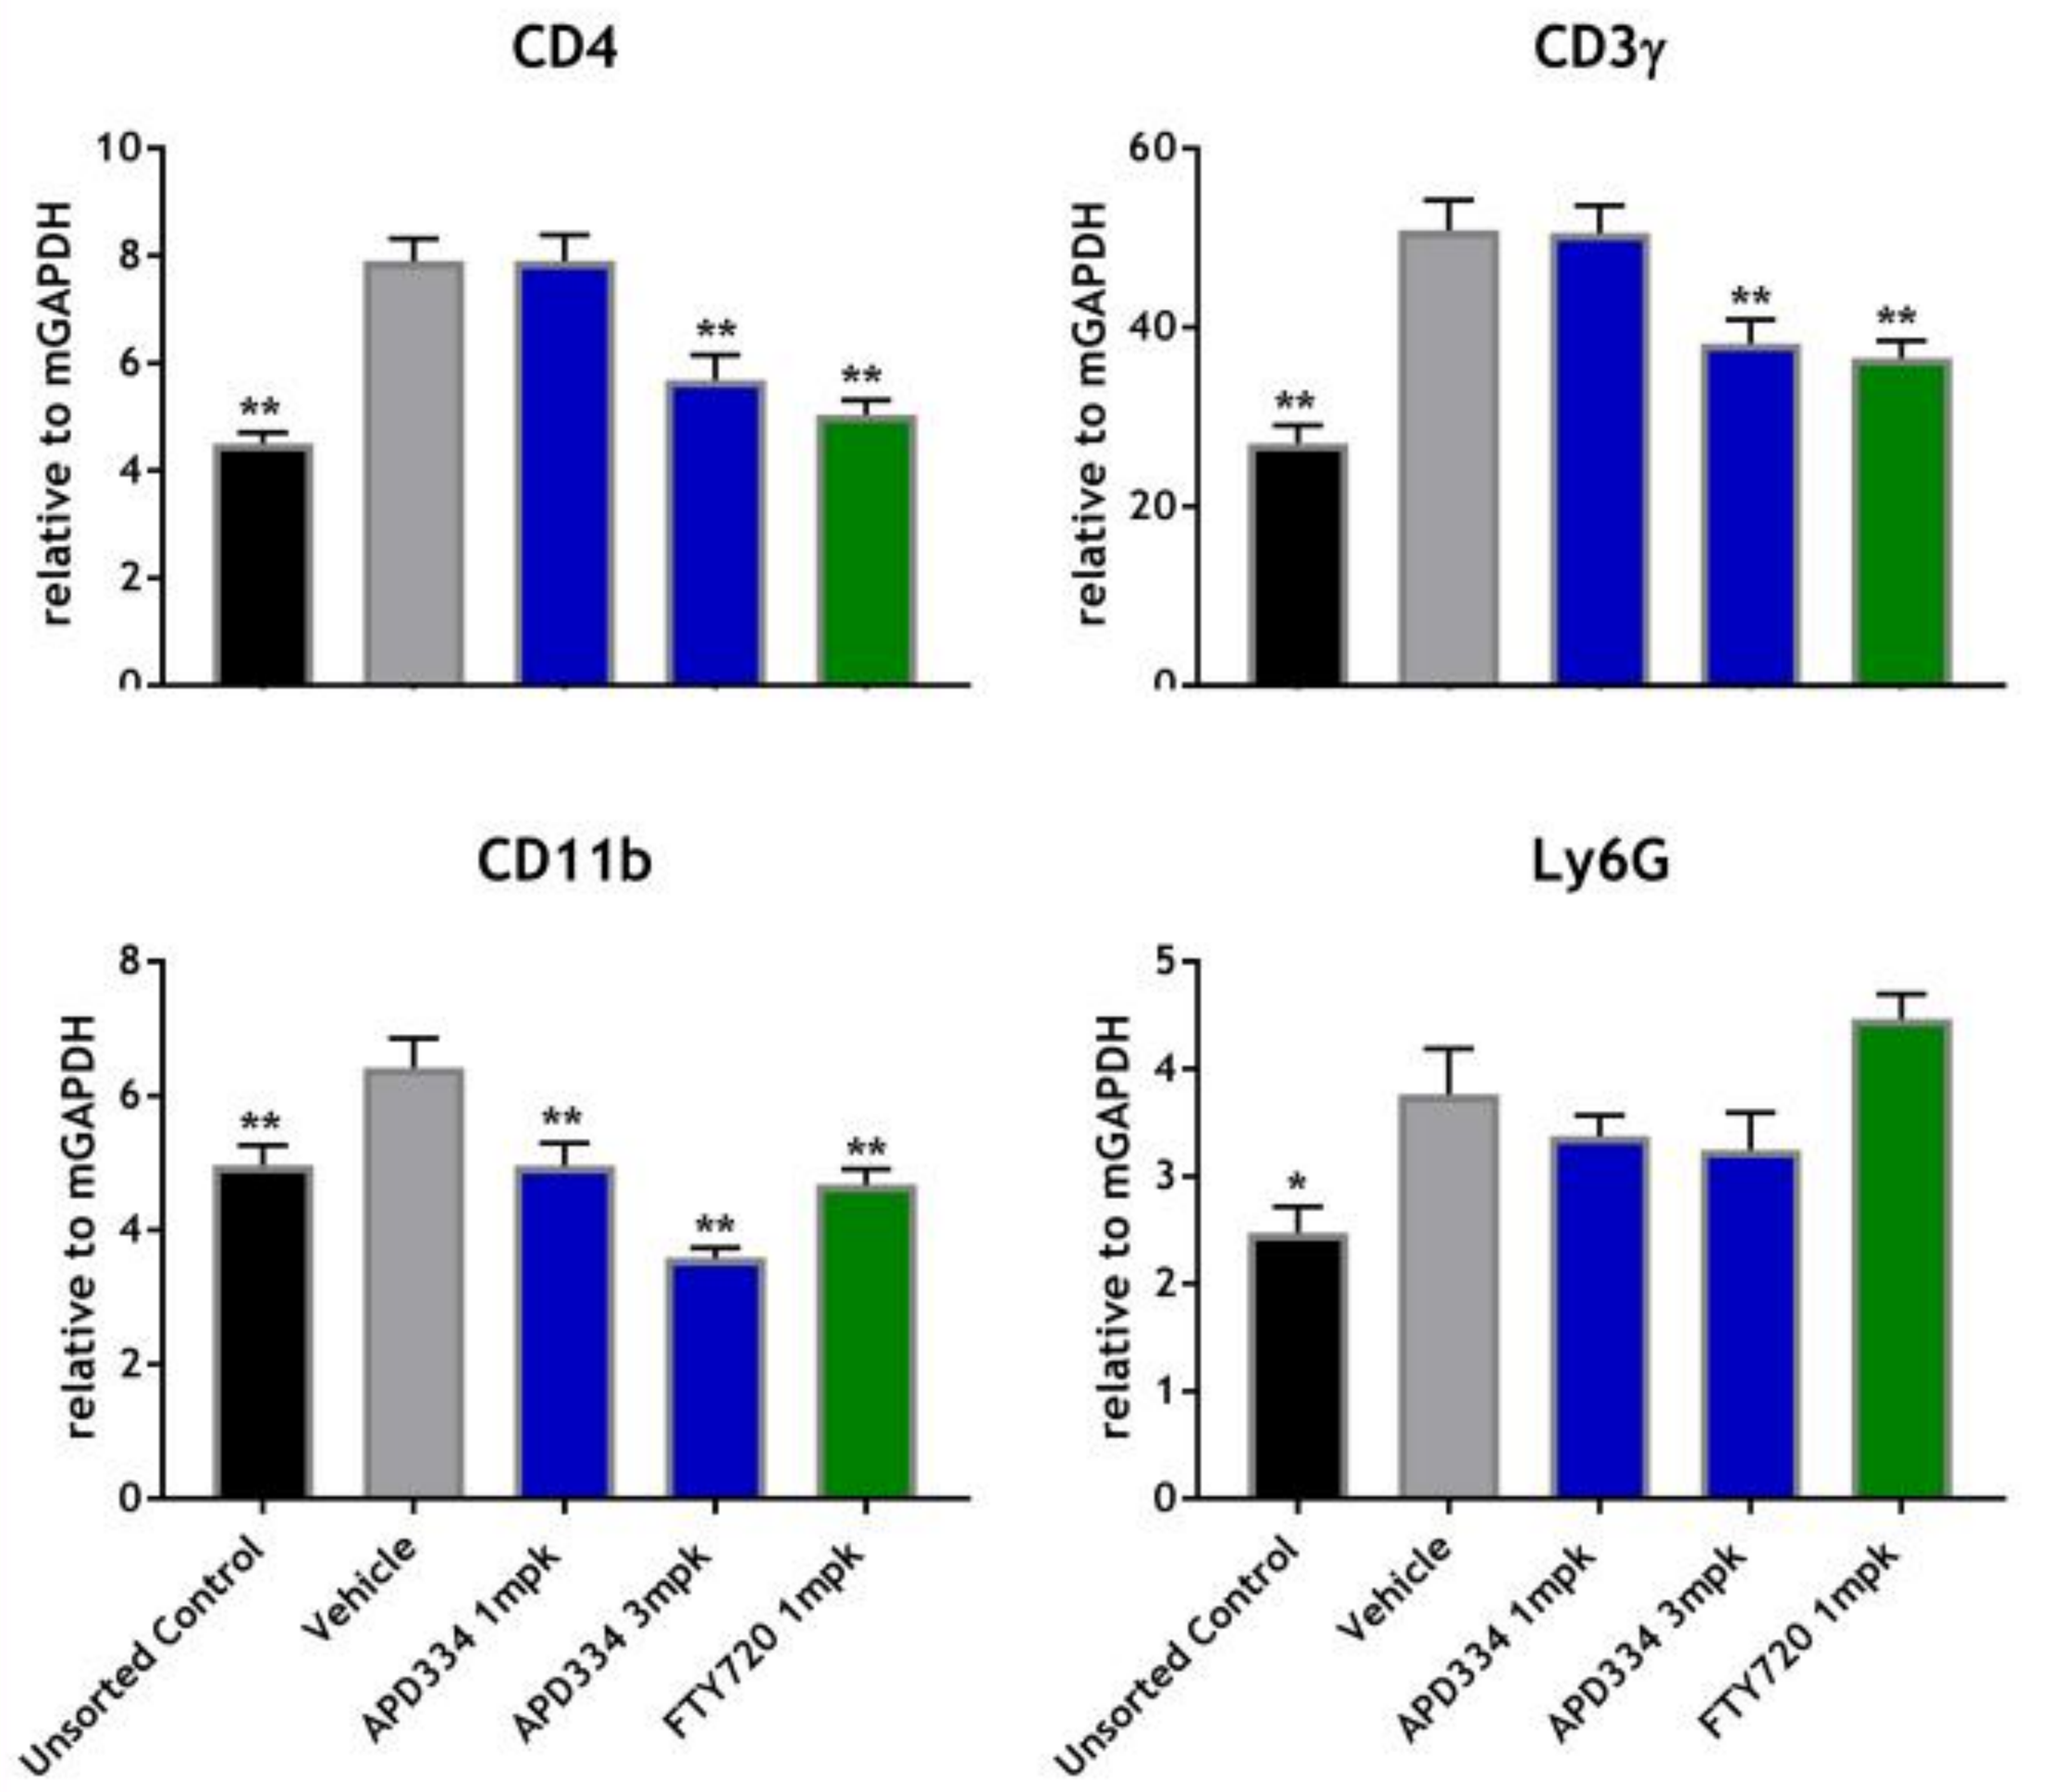
CD4
CD3γ
CD11b
Ly6G
relative to mGAPDH
Unsorted Control
Vehicle
APD334 1mpk
APD334 3mpk
FTY720 1mpk

Figure 3 *Etrasimod reduces infiltration and expansion of T cell and macrophage populations. qPCR of colon homogenates shows statistically significant decreases in CD4 (T cells and macrophages), CD3y (T cells) and CD11b (macrophages), but not Ly6G (neutrophils) in mice treated w/etrasimod. vs. vehicle *p<0.05; **p<0.01.*

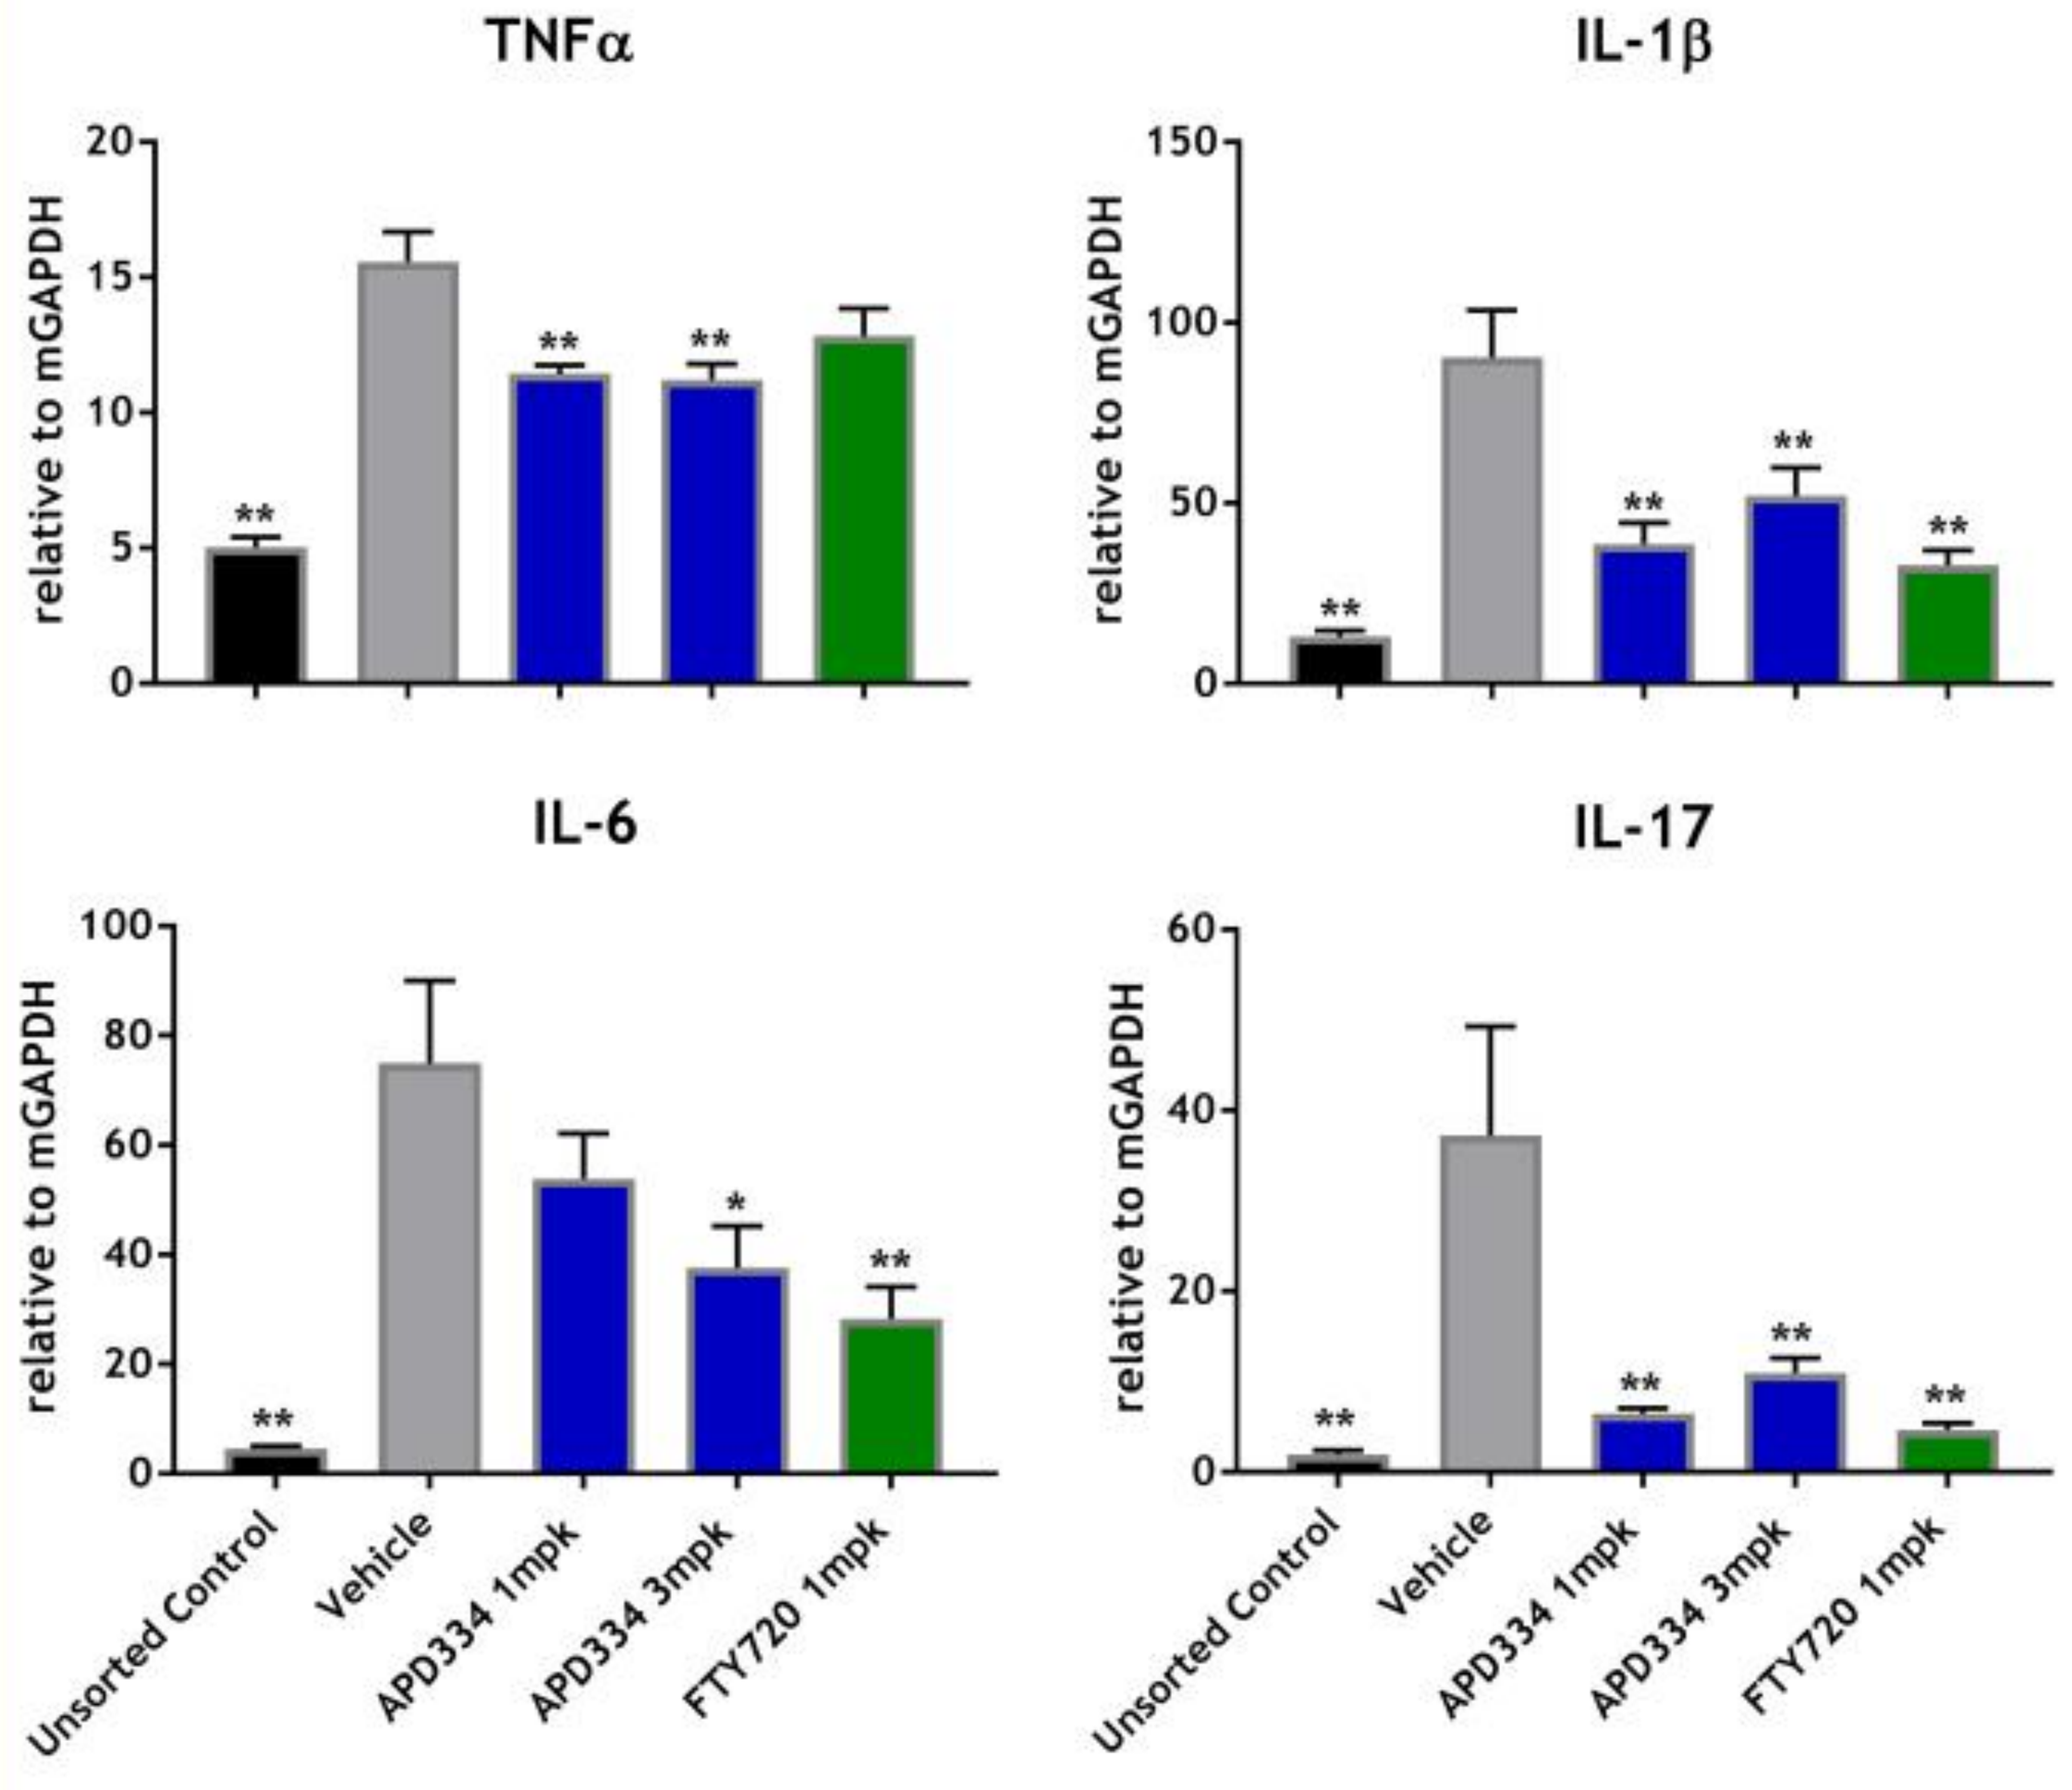
TNFα
IL-1β
IL-6
IL-17
relative to mGAPDH
Unsorted Control
Vehicle
APD334 1mpk
APD334 3mpk
FTY720 1mpk

Figure 4 *Etrasimod reduces elevations in pro-inflammatory cytokines. qPCR analysis of colon tissue homogenates show statistically significant decreases in macrophage derived pro-inflammatory cytokines TNFa, IL-1β, and IL-6, and T cell derived IL-17 in response to treatment with etrasimod compared to vehicle control. p<0.05; **p<0.01.*

Conclusion

Etrasimod, a potent selective orally bioavailable S1P agonist, inhibits experimental colitis induced by adoptive transfer of naive colitogenic CD4+CD45RB^{high} T cells into lymphoid-null SCID mice. These data suggest etrasimod blocks CD4+ T lymphocyte and macrophage activation and recruitment into colon. Based on these results, etrasimod may provide systemic and local immune cell modulation in the treatment of UC.

1. Daniel et al., *J. Immunol.* 2007;**178**: 2458-2468.
2. Fuji et al., *American Journal of Physiology*, 2006; **291**: G267–G274.
3. Mizushima et al., *Inflammatory Bowel Diseases*, 2004; **10**: 182–192,
4. Deguchi et al., *Oncology Reports*, 2006; **16**: 699–703.
5. Song et al., *JPET*, 2008; **324**: 276–283.

Funded by Arena Pharmaceuticals, San Diego, CA